Enhancing dendritic cell therapy with oncolytic adenovirus 3 coding for CD40L

Contact: sadia.zafar@helsinki.fi

Sadia Zafar[1], , Mikko Siurala[1,2] Suvi Sorsa[1,2], Otto Hemminki[1], Riikka Havunen[1], Siri Tähtinen[1], Simona Bramante[1], Lotta Vassilev[1], Hongjie Wang[3], Andre Lieber[3,4], Silvio Hemmi[5], Tanja de Gruijl[6], Anna Kanerva [1,7], Akseli Hemminki.[1,2,8]

[1]Cancer Gene Therapy Group, Department of Oncology, University of Helsinki, Helsinki, Finland,[2]TILT Biotherapeutics Ltd, Helsinki, Finland,[3]Division of Medical Genetics, University of Washington, Seattle, Washington, USA,[4]Department of Pathology, University of Washington, Seattle, Washington, USA,[5]Institute of Molecular Life Sciences, University of Zurich, Zurich, Switzerland,[6]VU University Medical Center, Amsterdam, Netherlands,[7]Department of Obstetrics and Gynecology, Helsinki University Central Hospital, Helsinki, Finland,[8]Helsinki University Comprehensive Cancer Center, Helsinki, Finland

Background

- ✓ Dendritic cell (DC) therapy is currently considered as a promising therapeutic option for treatment of cancers.
- ✓ However, tumor induced immunosuppression impairs the function of dendritic cells, eventually leading to tumor tolerance.
- ✓ Oncolytic adenoviruses cause biological effects which could be utilized to reverse the immunosuppression.

Aims

- ✓ To assess the antitumor efficacy of CD40L coding virus *in vivo* in combination with DC therapy in the presences of human immune cells.
- ✓ To study the correlation between the adenovirus expressing CD40L and DCs combination treatment with induction of antitumor immune response.

Results

1 Ad3-hTERT-CMV-hCD40L infected tumor cells induces DCs maturation, resulting T cell stimulation in a mixed lymphocyte reaction *ex vivo*

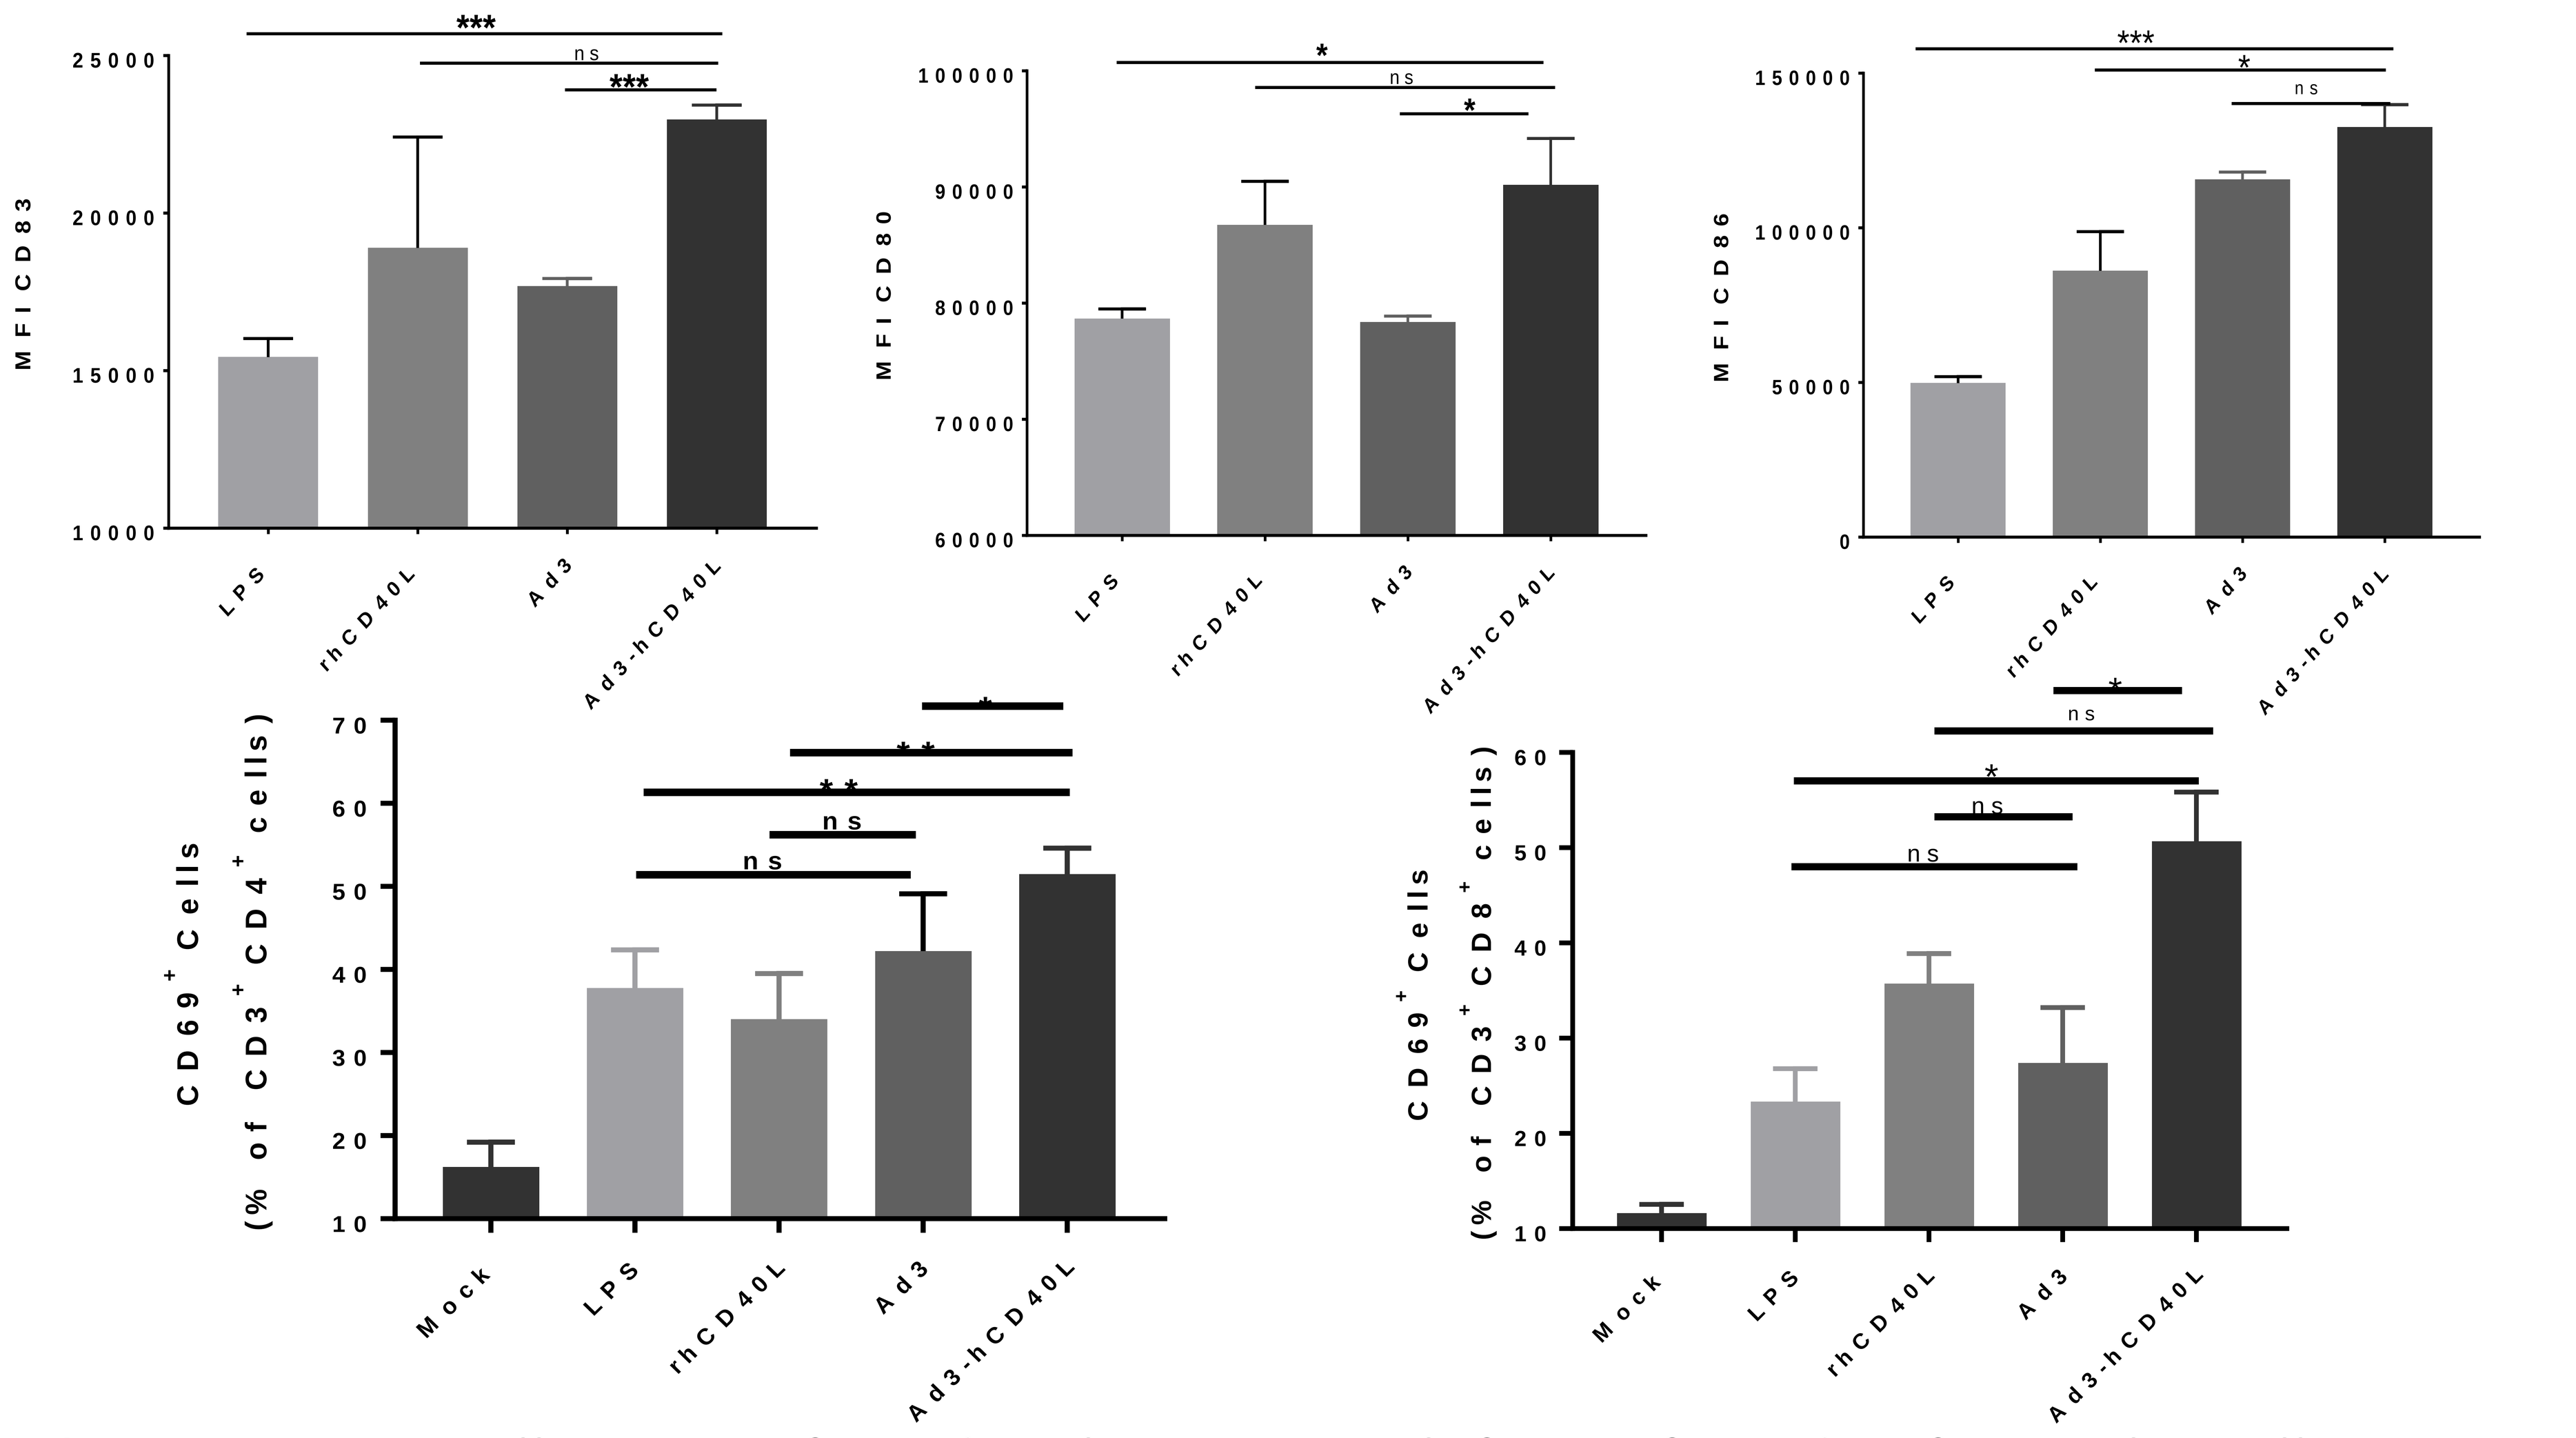
MFI CD83
MFI CD80
MFI CD86
CD69+ Cells (% of CD3+ CD4+ cells)
CD69+ Cells (% of CD3+ CD8+ cells)
Mock
LPS
rhCD40L
Ad3
Ad3-hCD40L

Figure 1: A549 cells were infected with viruses or left uninfected . After 18 h., cells were washed with PBS before adding immature dendritic cells. After 48h., co-culture DCs were assayed for maturation or co-cultured with T cells. . T cell activation was assessed 24hr later by flow cytometry.. LPS, lipopolysaccharide, rhCD40L: recombinant hCD40L protein, Ad3-hCD40L and Ad3: cells infection with Ad3-hTERT-CMV-hCD40L and Ad3-hTERT-E1A viruses, respectively.

2. Combination of Ad3-hTERT-CMV-hCD40L, DCs and human peripheral blood mononuclear cells PBMCs kill tumor cells *ex vivo*

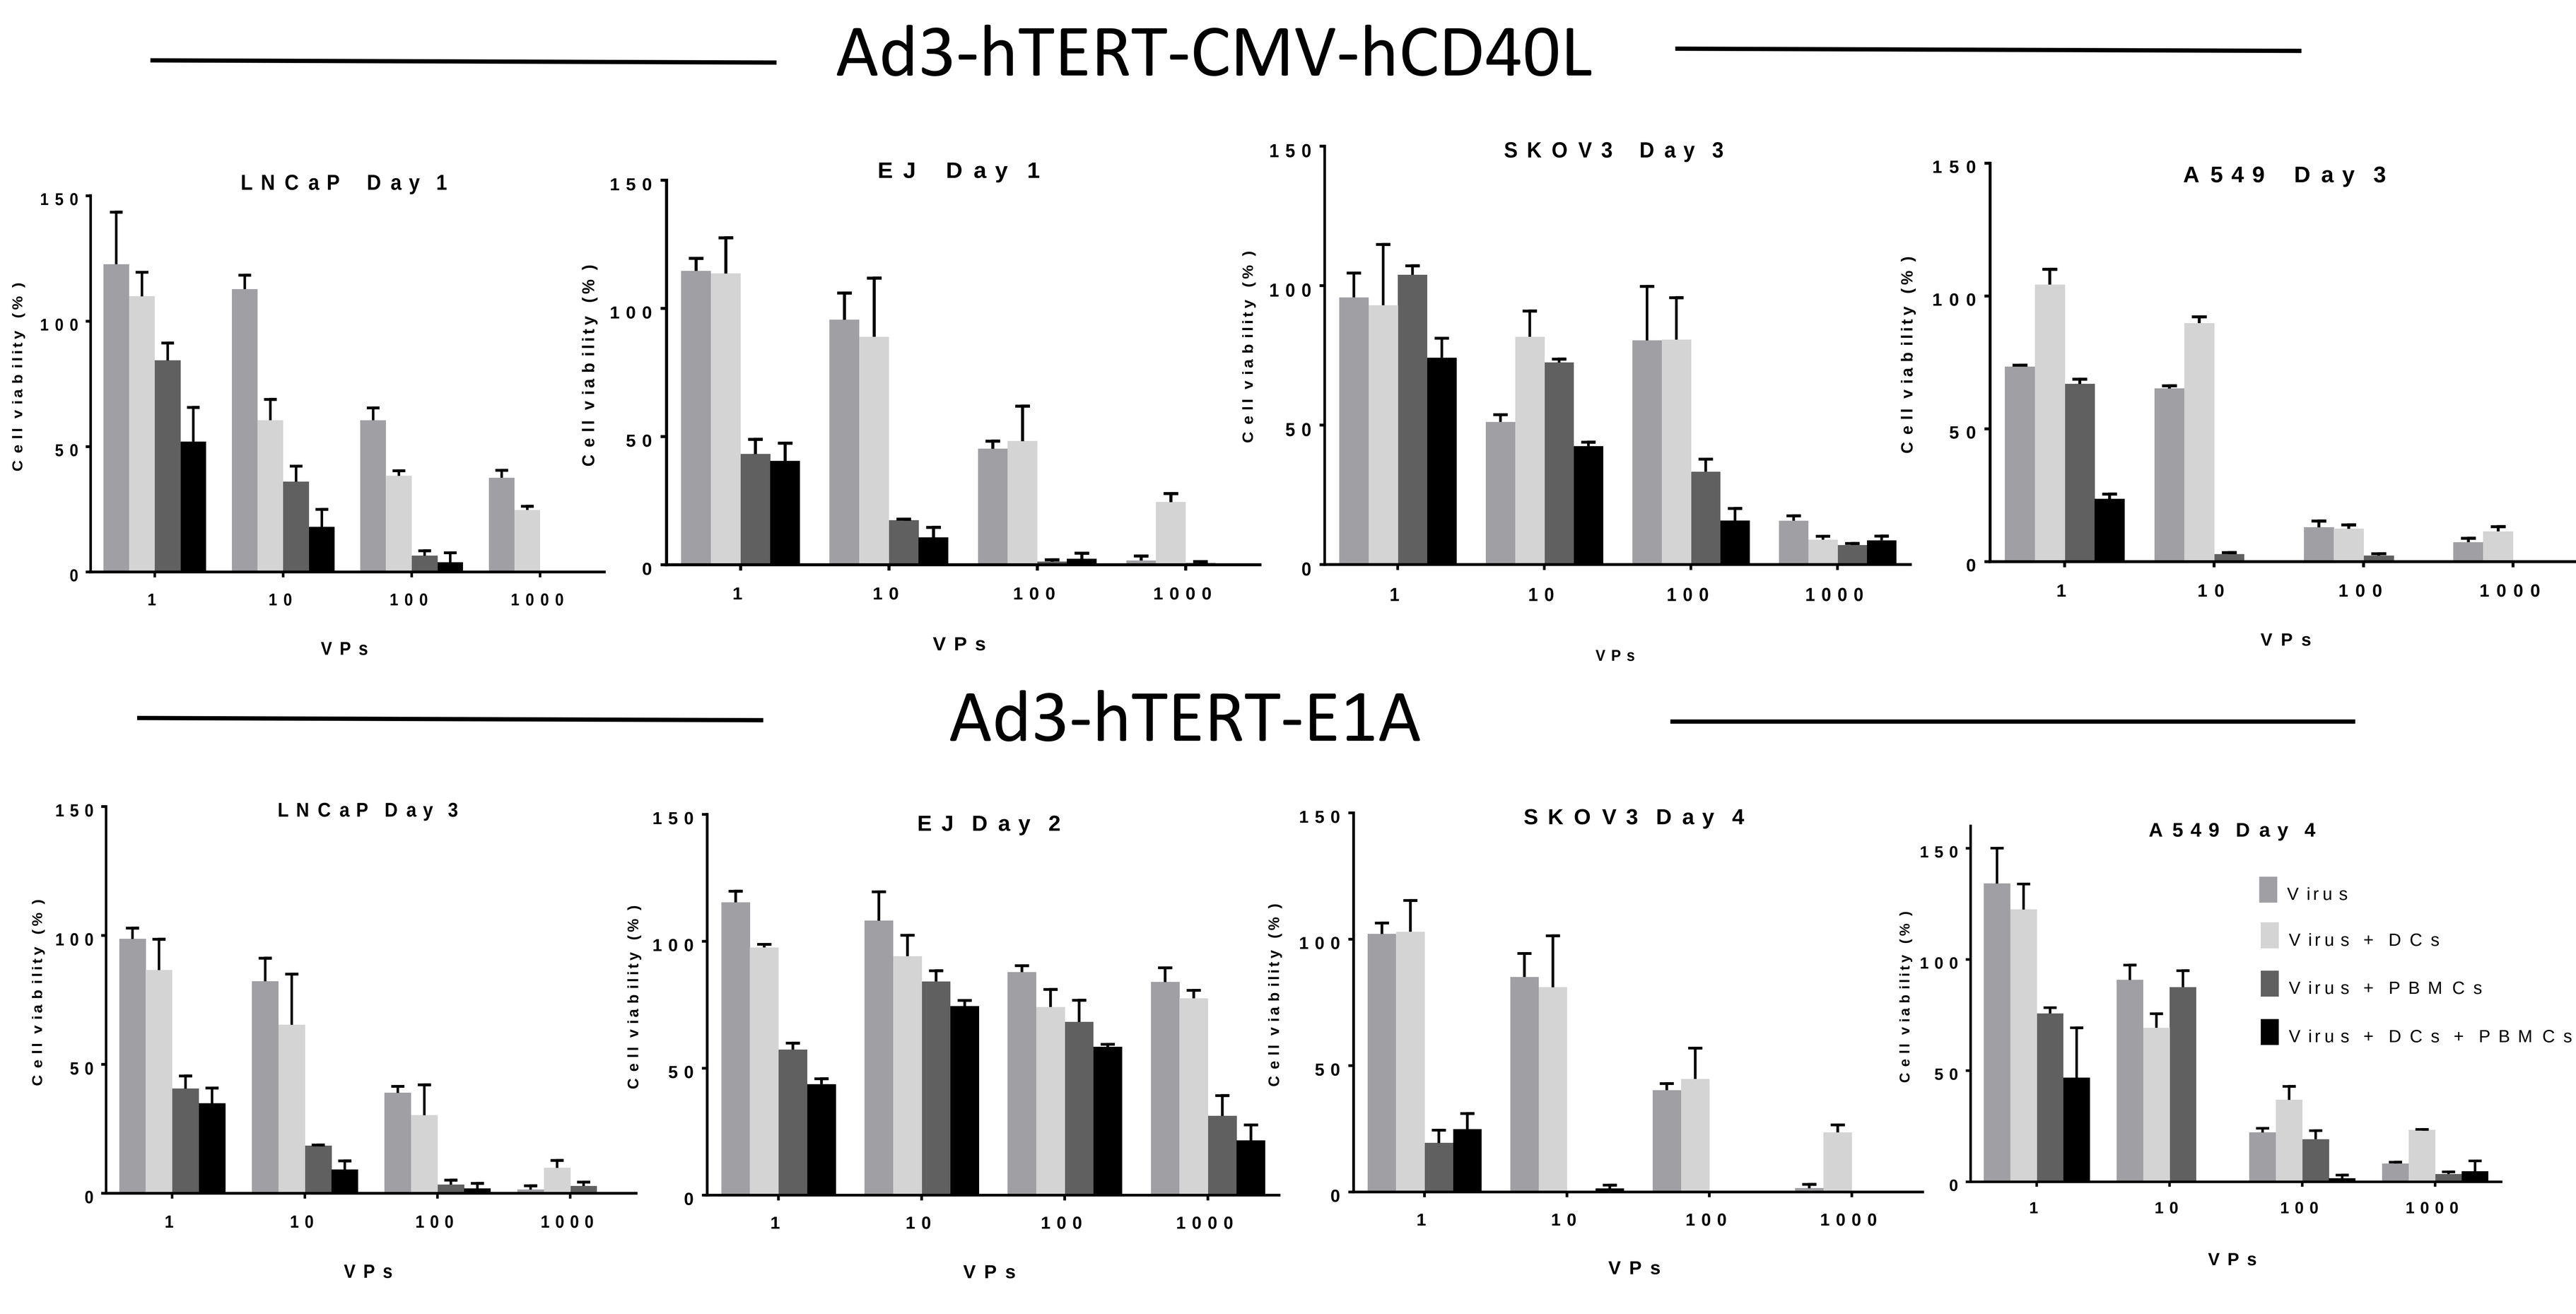
Ad3-hTERT-CMV-hCD40L
LNCaP Day 1
EJ Day 1
SKOV3 Day 3
A549 Day 3
Ad3-hTERT-E1A
LNCaP Day 3
EJ Day 2
SKOV3 Day 4
A549 Day 4
Cell viability (%)
VPs
Virus
Virus + DCs
Virus + PBMCs
Virus + DCs + PBMCs

Figure 2: Tumor killing potency of Ad3-hTERT-CMVhCD40L or Ad3-hTERT-E1A, DCs and PBMCs was assessed in LNCaP, EJ , SKOV3 and A549 cells. Cell killing was assessed at different time points (after adding DCs and PBMCs in co-culture) with MTS assay.

3. Antitumor efficacy of Ad3-hTERT-CMV-hCD40L and dendritic cell combination in a mice humanized with human peripheral blood mononuclear cells (PBMCs).

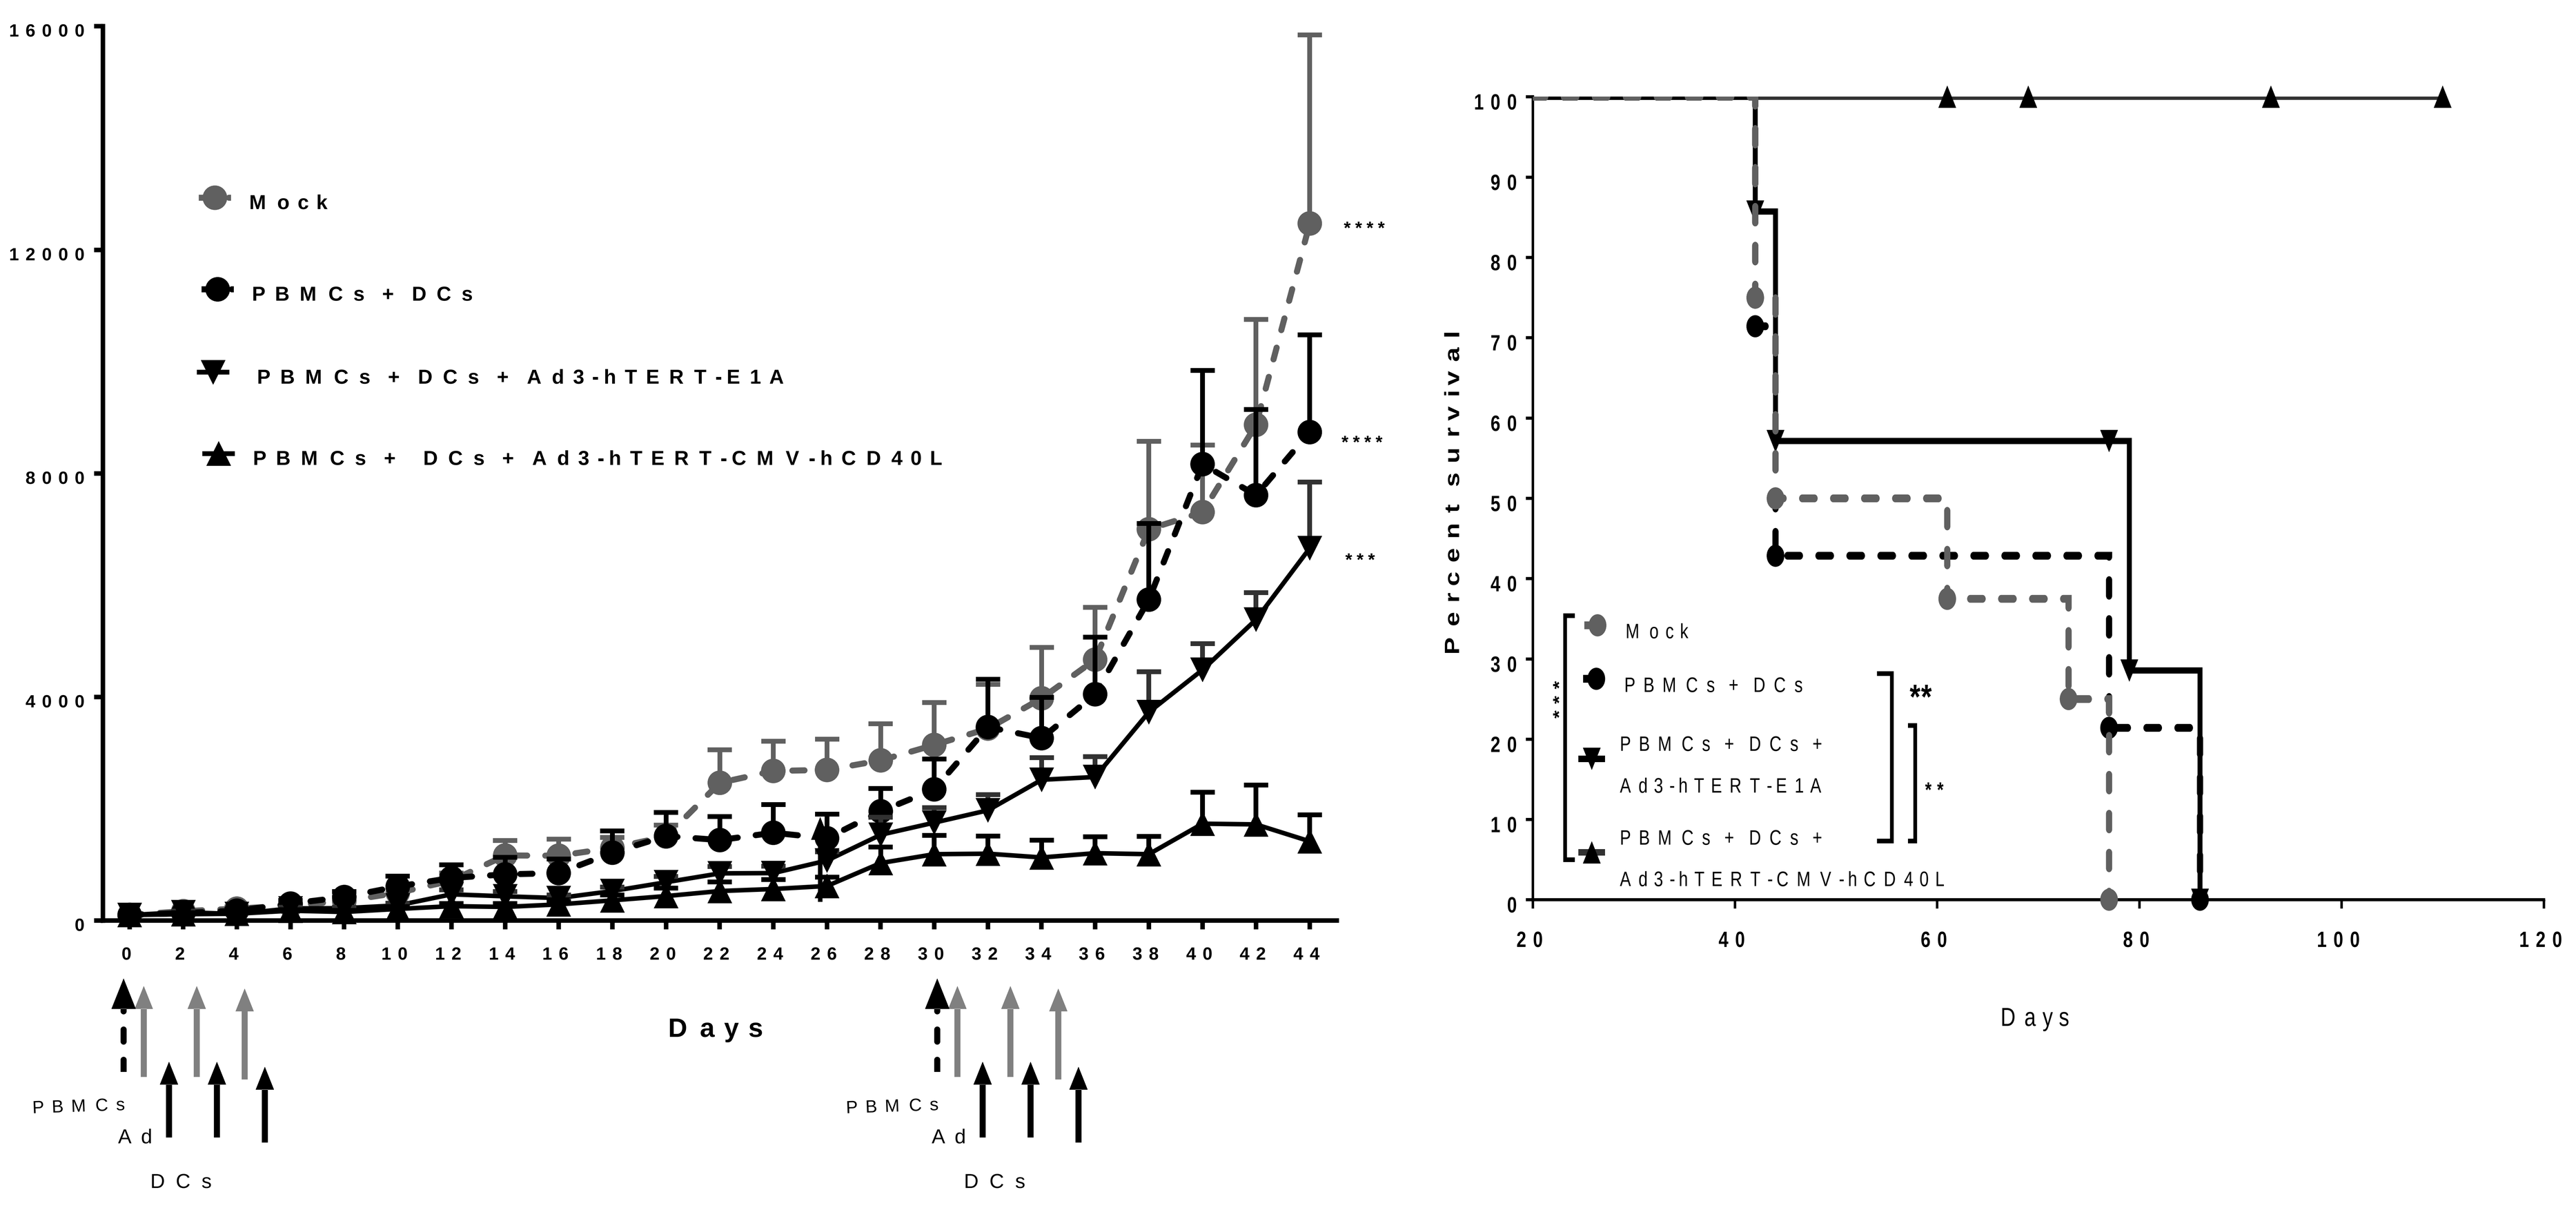
Mock
PBMCs + DCs
PBMCs + DCs + Ad3-hTERT-E1A
PBMCs + DCs + Ad3-hTERT-CMV-hCD40L
(Tumor growth %)
Days
PBMCs
Ad
DCs
Percent survival

Figure 3: A549 tumors were injected subcutaneously in immunodeficient mice. Viruses and dendritic cells were injected intratumorally three times. PBMCs were injected intravenously once. Tumor growth is expressed as percentage increase from first day of virus injection. Overall survival and statistic significances.

4. Combination of Adenovirus coding CD40L with DCs in mice humanized with PBMCs enhances anti-tumor immune response *in vivo* in tumors

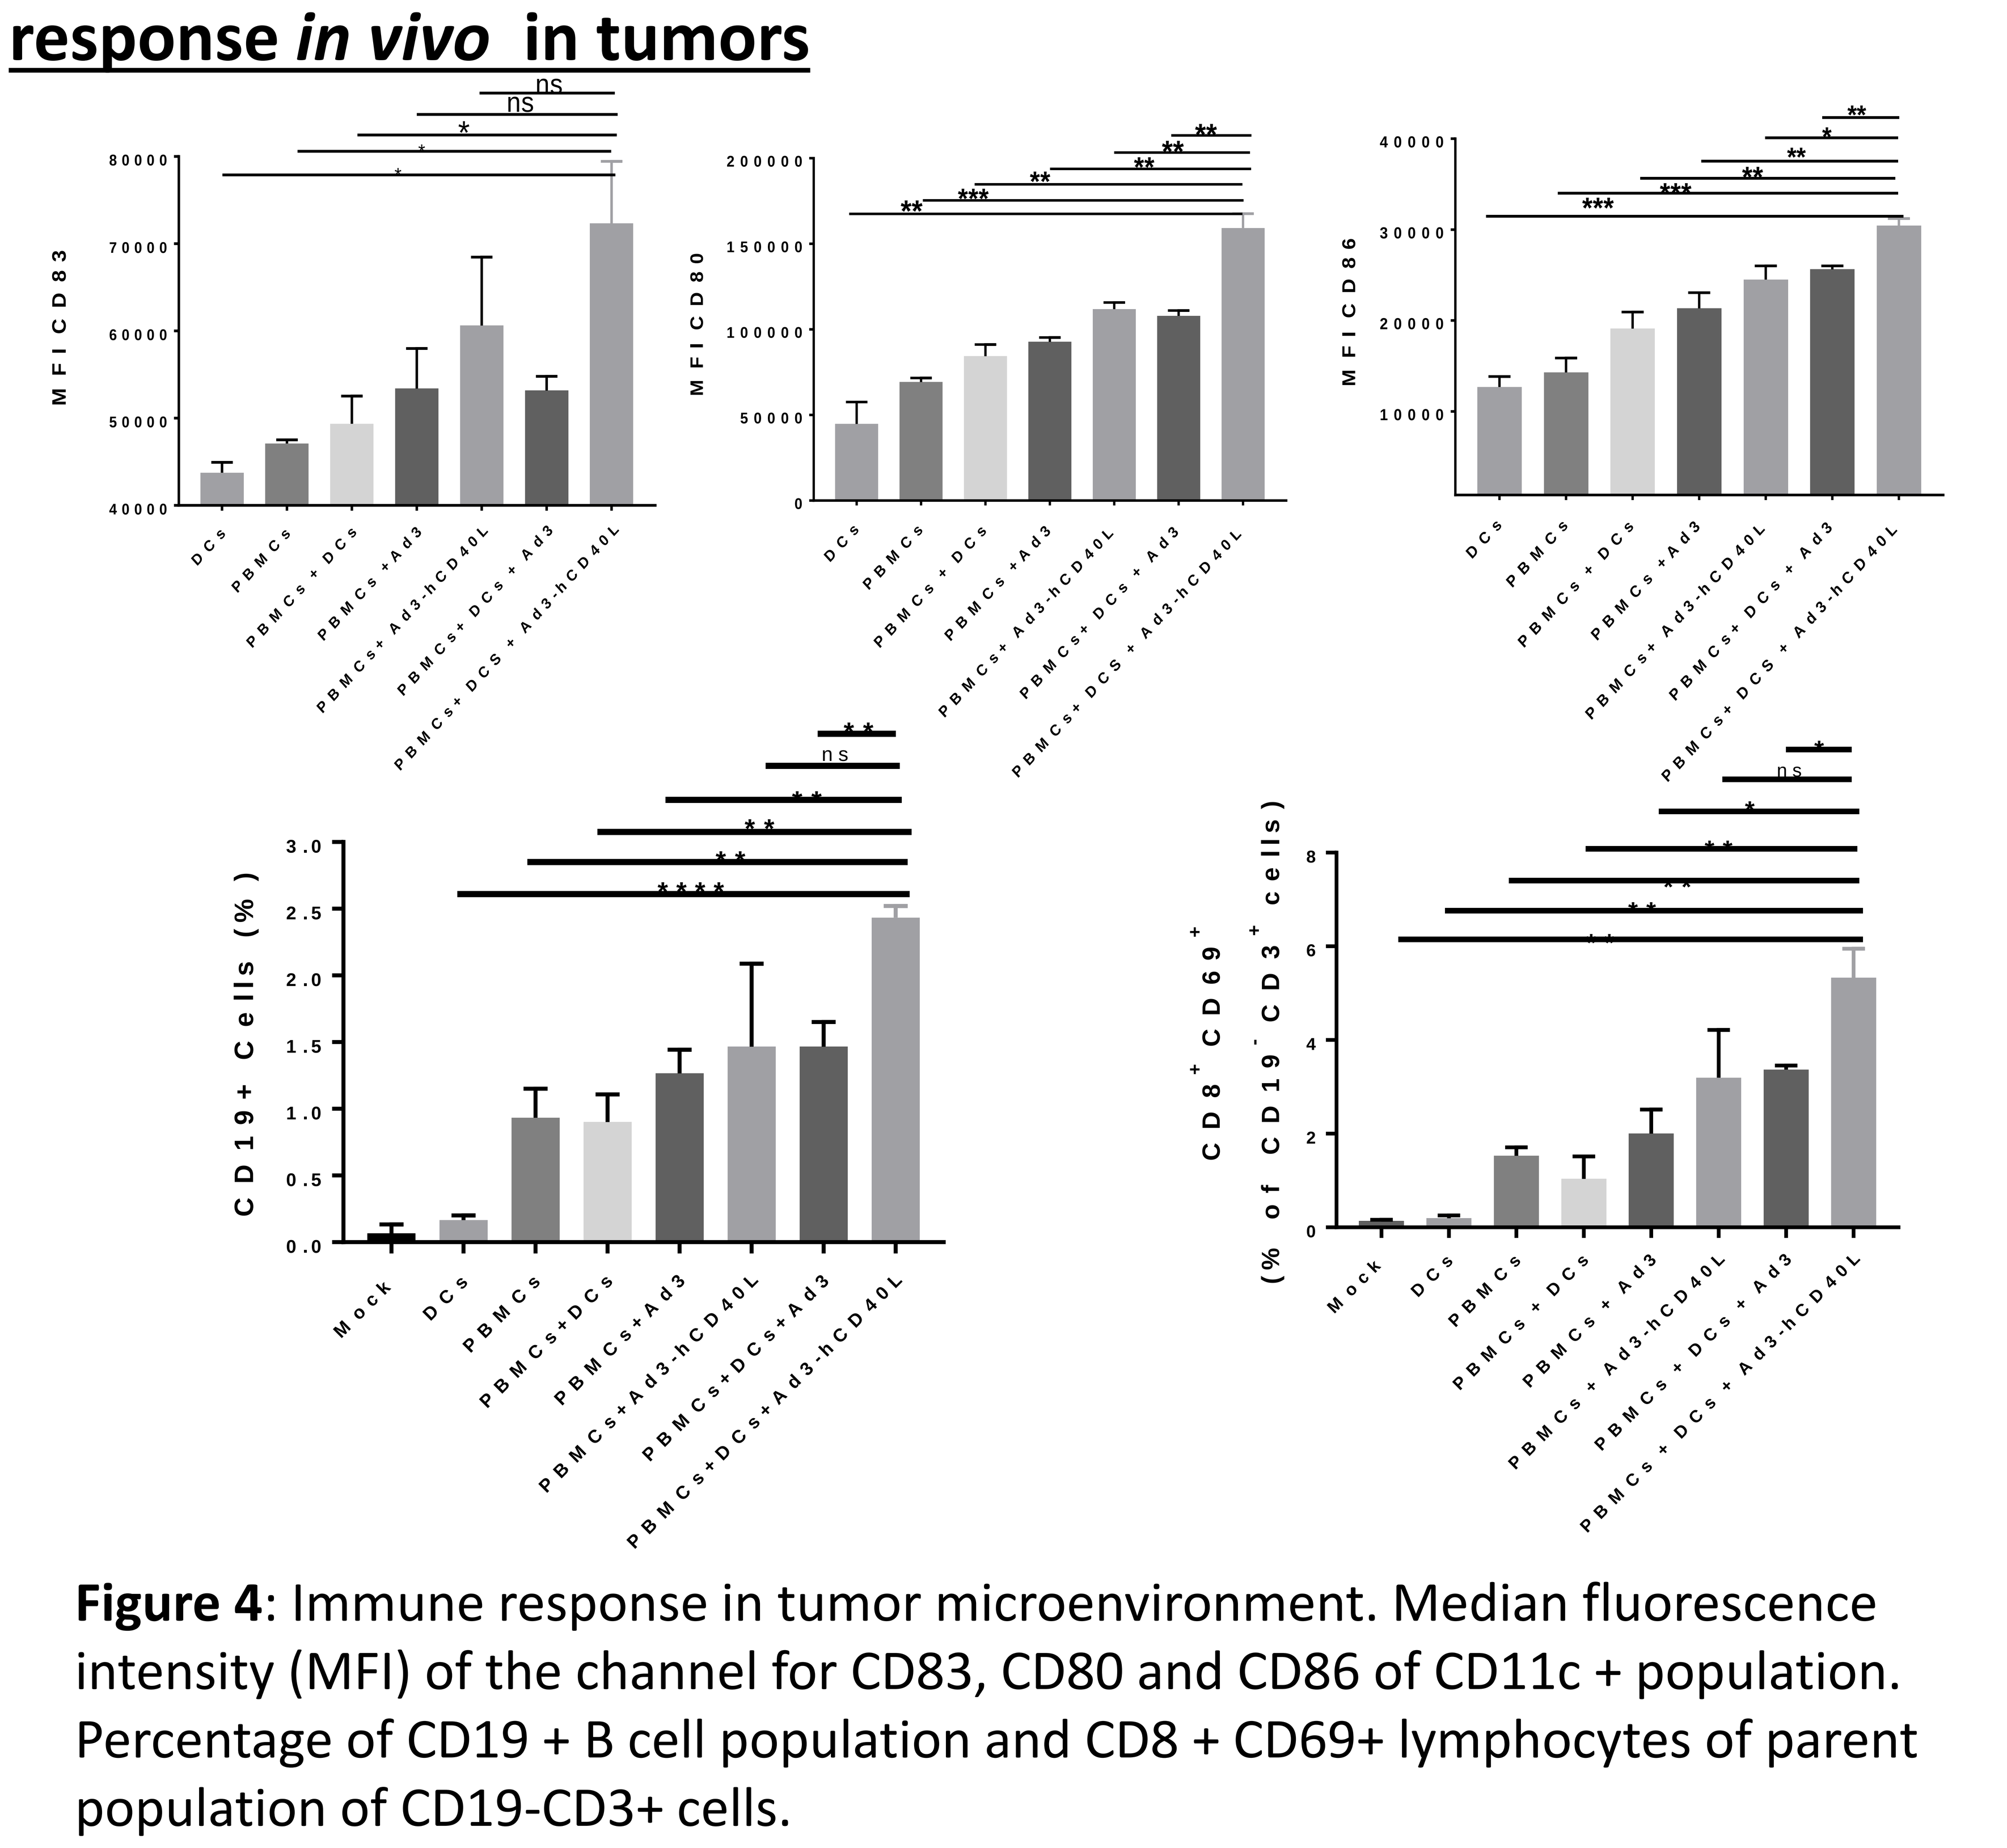
MFI CD83
MFI CD80
MFI CD86
CD19+ Cells (%)
CD8+ CD69+ (% of CD19- CD3+ cells)
Mock
DCs
PBMCs
PBMCs + DCs
PBMCs + Ad3
PBMCs + Ad3-hCD40L
PBMCs + DCs + Ad3
PBMCs + DCs + Ad3-hCD40L

Figure 4: Immune response in tumor microenvironment. Median fluorescence intensity (MFI) of the channel for CD83, CD80 and CD86 of CD11c + population. Percentage of CD19 + B cell population and CD8 + CD69+ lymphocytes of parent population of CD19-CD3+ cells.

Conclusions

- ✓ Ad3-hTERT-CMV-hCD40L infected tumor cells induces maturation of human DCs which in turn required for T cell activation *ex vivo* and *in vivo*.
- ✓ We have shown promising anti-tumor immune responses suggesting that adenovirus coding for CD40L successfully enables dendritic cell therapy.
- ✓ Ad3-hTERT-CMV-hCD40L is promising for translation into human trials. In particular, this virus could enable successful dendritic cell therapy in cancer patients.